1822P

Incidence of cachexia and health care resource use (HCRU) in patients with breast, colorectal, lung, pancreatic, and prostate cancers

Imran Ali[1], Simon Dagenais[2], Ira Jacobs[2], Shelby Corman[3], Taiwo Adesoba[3], Feng Dai[2], Oluwaseyi Dina[2]

[1]Icahn School of Medicine at Mount Sinai, New York, NY, USA;
[2]Pfizer Inc, New York, NY, USA; [3]Precision AQ™, Bethesda, MD, USA

Introduction

- Cachexia in patients with cancer is characterized by involuntary weight loss, muscle atrophy, increased lipolysis and energy expenditure, and reduced appetite.[1]
- It can lead to a decreased quality of life, reduced cancer treatment effectiveness, increased susceptibility to cancer treatment side effects, and increased cancer-related mortality.[2,3]
- Based on available scientific literature from clinical studies, cachexia affects ~50–80% of patients with advanced cancer and ~20% of cancer deaths are attributable to cachexia.[3,4]
- Cachexia is likely to be under-diagnosed due to inconsistent application of diagnostic criteria in clinical practice as well as challenges when interpreting changes in bodyweight (for example, due to obesity, edema, and/or ascities).[5,6]
- There are also limited data on how cachexia may impact health care resource use (HCRU) in patients with cancer.[7]

Objectives

- To estimate the incidence of cachexia in patients newly diagnosed with breast, colorectal, lung, pancreatic, and prostate cancer.
- To compare measures of health care resource utilization in patients diagnosed with cancer who do and do not have cachexia.

Methods

- This retrospective, observational, cohort study integrated electronic health record and administrative claims data using the Optum® Market Clarity database (**Figure 1**).

Figure 1: Study design

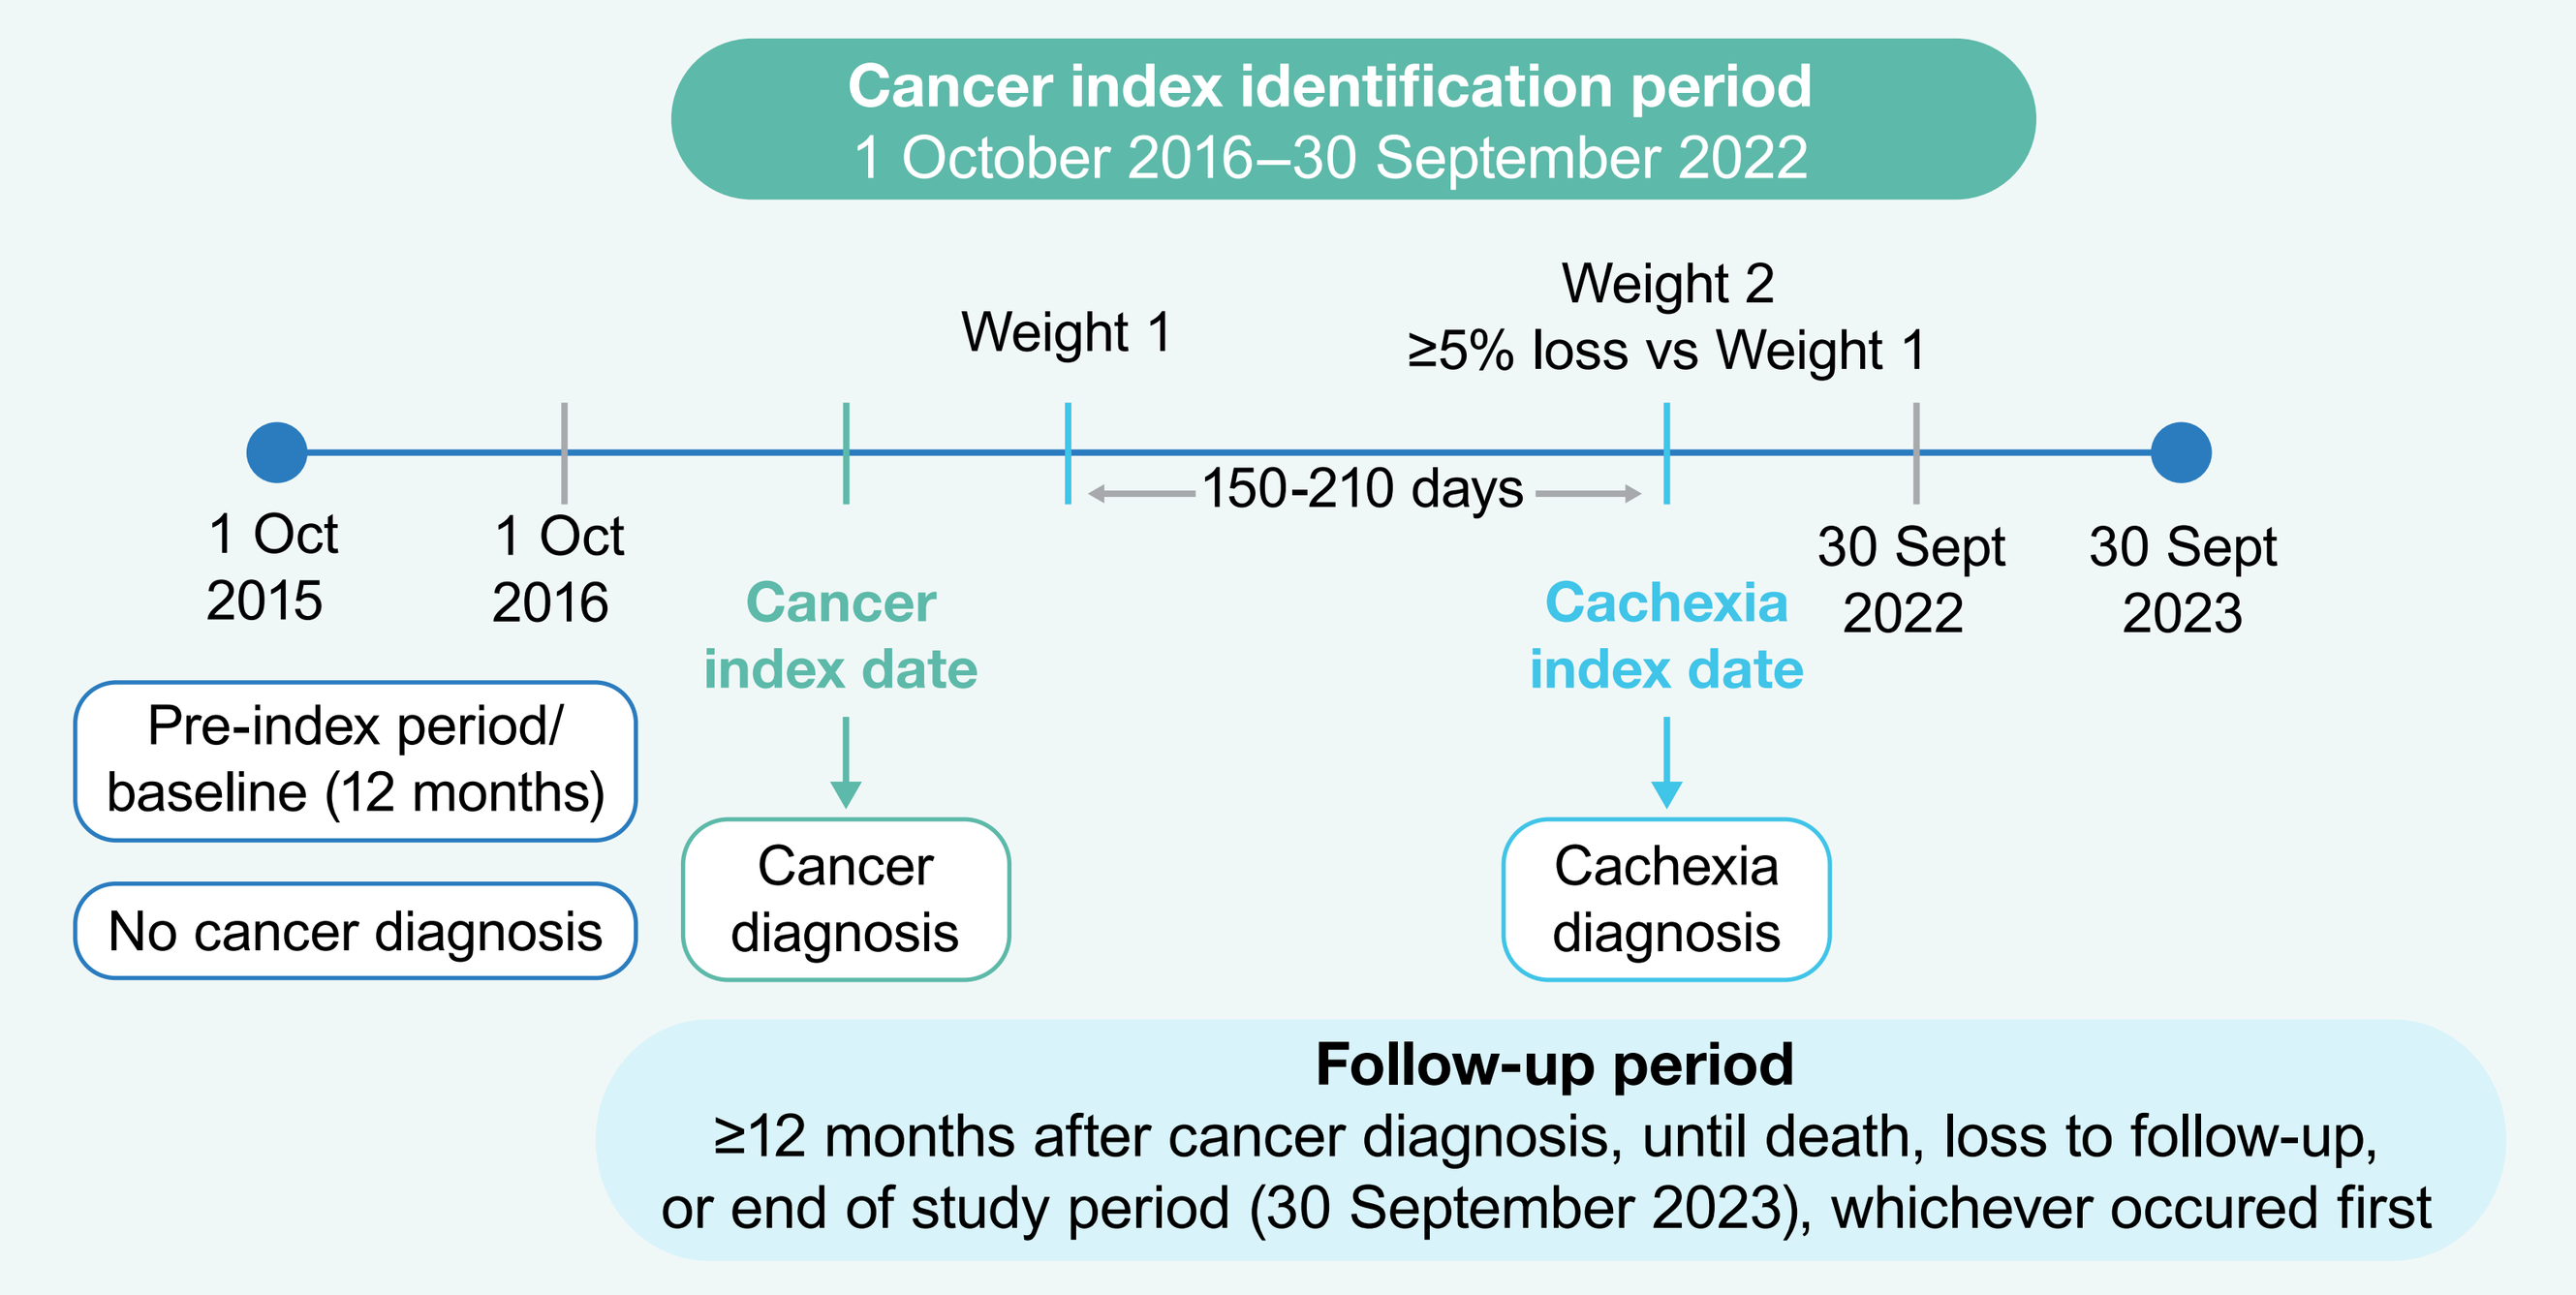
Cancer index identification period
1 October 2016–30 September 2022
Weight 1
Weight 2
≥5% loss vs Weight 1
150-210 days
1 Oct 2015
1 Oct 2016
Cancer index date
Cachexia index date
30 Sept 2022
30 Sept 2023
Pre-index period/ baseline (12 months)
No cancer diagnosis
Cancer diagnosis
Cachexia diagnosis
Follow-up period
≥12 months after cancer diagnosis, until death, loss to follow-up, or end of study period (30 September 2023), whichever occured first

- Inclusion and exclusion criteria are shown in **Table 1**.
- Cachexia was defined as ≥5% loss in bodyweight calculated from 2 bodyweight measurements ~6 months apart.[8]
- The cachexia index date was the earliest date after the cancer index date on which ≥5% loss of bodyweight was recorded.
- In the control group, the cachexia index date was the date after the cancer index date on which <5% loss of bodyweight was recorded.
- The primary outcome was the number of health care visits per-patient-per-month (PPPM; 30 days) during the 12 months post-cachexia index date.
- Inverse probability of treatment weighting was used to adjust for confounding between cohorts (cachexia vs no cachexia) at the cachexia index date.
- Student's *t*-test was used to compare HCRU between cohorts (cachexia vs no cachexia) by tumor type.

Table 1: Eligibility criteria

Inclusion criteria

- Diagnosed with ≥1 malignant solid tumor of interest between 1 October 2016 and 30 September 2022, as identified by the following ICD-10-CM codes in ≥1 claim or encounter in a hospital inpatient setting, or ≥2 claims or encounters in selected outpatient setting ≥30 days apart:
 - Breast cancer (ICD-10 codes C50.x)
 - Colorectal cancer (ICD-10 codes C18.x-C21.x, C49.x, C7A.x)
 - Lung cancer (ICD-10 codes C34.x, C7A.090)
 - Pancreatic cancer (ICD-10 codes C25.x)
 - Prostate cancer (ICD-10 codes C61.x)
- ≥19 years of age at cancer index date (ie, ≥18 years of age at the beginning of the 12 months before cancer index date)
- EHR enrollment (ie, EHR activity) for 12 months before cancer index date
- ≥2 bodyweight assessments 150–210 days apart in the 12 months before cancer index
- ≥2 bodyweight assessments 150–210 days apart with ≥1 of those assessments occurring in the 12 months after cancer index
- EHR enrollment for 12 months after cancer index date or death, whichever occured first

For analyses related to HCRU:

- Continuous health plan enrollment for 12 months before cachexia index date
- Continuous health plan enrollment for ≥1 month after cachexia index or until death, whichever occured first

Exclusion criteria

- In the 12 months before the cancer index date, ≥1 claim or encounter in any setting with a diagnosis of:
 - Any solid tumor other than the index (incident cancer) diagnosis (ICD-10 codes C00.x-C75.x or C7A.x)
 - Any hematologic malignancy (ICD-10 codes C81.x-C96.x)
 - Any cancer with metastasis (ICD-10 codes C77.x, C78.x, C79.x, C7B.x)
 - Any other cancer type (ICD-10 codes C45.x, C46.x, C76.x, C80.x, C96.x)
- History of cachexia in the pre-index period (ie, patient met criteria for cachexia in the 12 months before the cancer index date, as defined below) but not in the post-index period:
 - 2 bodyweight measurements within 150–210 days of each other
 - 2nd bodyweight was ≤95% of the first bodyweight measurement
- Unknown gender
- Prostate cancer cohort only: Female gender

EHR=electronic health record; HCRU=health care resource use; ICD-10-CM=International Classification of Diseases, 10th Revision, Clinical Modification

Results

INCIDENCE

- A total of 27,428 patients with a malignant solid tumor were included.
- Within 12 months of receiving a cancer diagnosis, cachexia was observed in 38% of patients.

BREAST CANCER

Population size: 10,500
Incidence of cachexia: 34%

Demographics	With cachexia n=3595	Without cachexia n=6905	P value
Female, %	99	99	0.31
Age, mean (SD), y	65 (12)	65 (12)	0.74
Race, %			0.87
African American	13	12	
Asian	2	2	
White	81	82	
Other/Unknown	4	4	

HCRU	With cachexia n=3595	Without cachexia n=6905	P value
Inpatient	0.98	0.80	**0.05**
Outpatient	5.11	3.46	**<0.0001**
Office	3.69	3.44	**0.02**
Home	0.67	0.83	**0.05**
Telehealth	0.03	0.03	0.51
Other	4.75	3.85	**<0.0001**
Overall	15.22	12.41	**<0.0001**

COLORECTAL CANCER

Population size: 3304
Incidence of cachexia: 51%

Demographics	With cachexia n=1683	Without cachexia n=1621	P value
Female, %	52	52	0.92
Age, mean (SD), y	66 (13)	66 (13)	0.83
Race, %			0.98
African American	11	11	
Asian	1	1	
White	83	83	
Other/Unknown	5	5	

HCRU	With cachexia n=1683	Without cachexia n=1621	P value
Inpatient	2.72	2.32	0.21
Outpatient	5.36	3.93	**<0.0001**
Office	4.38	3.55	**<0.001**
Home	1.51	0.84	**<0.0001**
Telehealth	0.02	0.02	0.37
Other	6.49	5.63	0.07
Overall	20.48	16.30	**<0.0001**

LUNG CANCER

Population size: 3345
Incidence of cachexia: 55%

Demographics	With cachexia n=1845	Without cachexia n=1500	P value
Female, %	57	56	0.55
Age, mean (SD), y	69 (10)	69 (10)	0.94
Race, %			0.93
African American	10	10	
Asian	1	1	
White	86	86	
Other/Unknown	3	3	

HCRU	With cachexia n=1845	Without cachexia n=1500	P value
Inpatient	6.92	5.45	**0.03**
Outpatient	6.89	6.14	0.07
Office	4.36	4.29	0.78
Home	1.89	1.89	1.00
Telehealth	0.02	0.02	0.74
Other	9.52	7.53	**<0.001**
Overall	29.59	25.32	**<0.001**

HEALTH CARE RESOURCE USE

- Across all tumor types, hospital outpatient and office visits PPPM were numerically greater in patients with vs without cachexia, with statistical significance varying by tumor type.
- Other differences in hospital inpatient, home, telehealth, and other visits were also observed.

PANCREATIC CANCER

Population size: 475
Incidence of cachexia: 74%

Demographics	With cachexia n=350	Without cachexia n=125	P value
Female, %	52	54	0.72
Age, mean (SD), y	67 (12)	67 (13)	0.90
Race, %			0.95
African American	15	17	
Asian	1	1	
White	79	79	
Other/Unknown	5	4	

HCRU	With cachexia n=350	Without cachexia n=125	P value
Inpatient	8.25	12.50	0.13
Outpatient	10.80	8.72	0.18
Office	6.41	2.66	**<0.0001**
Home	2.49	1.47	0.09
Telehealth	0.05	0.02	0.21
Other	12.60	14.08	0.58
Overall	40.59	39.45	0.83

PROSTATE CANCER

Population size: 9804
Incidence of cachexia: 29%

Demographics	With cachexia n=2865	Without cachexia n=6939	P value
Male, %	100	100	–
Age, mean (SD), y	69 (9)	69 (9)	0.25
Race, %			0.94
African American	15	15	
Asian	1	1	
White	79	80	
Other/Unknown	4	4	

HCRU	With cachexia n=2865	Without cachexia n=6939	P value
Inpatient	1.46	0.69	**<0.0001**
Outpatient	3.14	2.50	**<0.0001**
Office	3.27	3.06	**0.03**
Home	0.64	0.43	**<0.01**
Telehealth	0.02	0.01	0.15
Other	4.53	3.20	**<0.0001**
Overall	13.04	9.89	**<0.0001**

CONCLUSIONS

- **Incidence of cachexia was highest in patients with pancreatic cancer followed by lung and colorectal cancer, and lowest in breast and prostate cancers.**
- **Patients with cachexia used more health care resources, highlighting a high clinical and economic burden in this population.**

Please scan this Quick Response (QR) code with your smartphone app to view this poster. If you do not have a smartphone, access the poster via the internet at: https://scientificpubs.congressposter.com/p/yy12t90sbkyhbzgt
Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

REFERENCES

1. Nishie K, et al. *Drug Discov Today* 2023;28:103689. 2. Dev R. *Ann Palliat Med* 2019;8:24-32. 3. von Haehling S, Anker SD. *J Cachexia Sarcopenia Muscle* 2014:261-3. 4. Argilés JM, et al. *Nat Rev Cancer* 2014;14:754-62. 5. Nishikawa H, et al. *Int J Mol Sci* 2021;22. 6. Bianchini C, et al. *Cancer Treat Rev* 2024;125:102717. 7. Tarricone R, et al. *Crit Rev Oncol Hematol* 2016;99:49-62. 8. Fearon K, et al. *Lancet Oncol* 2011;12:489-95.

ACKNOWLEDGMENTS

This study was undertaken by PRECISIONheor and sponsored by Pfizer Inc. Medical writing support was provided by Simon R. Stones, PhD, ISMPP CMPP™, of Engage Scientific Solutions Ltd, and funded by Pfizer Inc.
Contact: Shelby Corman, Shelby.Corman@precisionaq.com

DISCLOSURES

IA: honoraria and speakers' bureau: Pfizer; stock and other ownership interests: Eli Lilly, Merck, Novo Nordisk. **SD:** employment: Pfizer Inc; shares: Pfizer Inc. **IJ:** employment: Pfizer Inc; shares: Pfizer Inc. **SC**: Precision AQ™. **TA**: employment: Precision AQ™. **FD**: employment: Pfizer Inc; shares: Pfizer Inc. **OD**: employment: Pfizer Inc; shares: Pfizer Inc.

Presented at the European Society for Medical Oncology (ESMO) Congress, 13–17 September 2024, Barcelona, Spain

Copyright ©2024